Ovarian Activity in Obese and Non-Obese Women Treated With AG200-15, a Transdermal Contraceptive

Marie Foegh, MD,[1] David F. Archer, MD,[2] and Daniel R. Mishell Jr., MD[3]

[1]Agile Therapeutics, Princeton, New Jersey; [2]Jones Institute for Reproductive Medicine, Eastern Virginia Medical School, Norfolk, Virginia; [3]Keck School of Medicine, University of Southern California, Los Angeles, California

Abstract

Objective: Evaluate ovarian suppression in obese and non-obese healthy women treated with AG200-15, a weekly transdermal contraceptive delivery system (TCDS) containing levonorgestrel and ethinyl estradiol.

Methods: Luteal activity in obese (BMI at least 30 kg/m^2, n=8) versus non-obese (BMI below 30 kg/m^2, n=25) women was compared using data from a previously reported multicenter, open-label, randomized, 3-cycle study evaluating AG200-15, a weekly TCDS (21/7 day cycle). Luteal activity (defined as serum-progesterone [s-P] 3 ng/mL or more) was measured twice weekly. The cycle day of increased s-P was determined.

Results: BMI ranged from 17-52 kg/m^2; 24% of subjects were obese. Progesterone values, available in 30 women, revealed no differences in ovarian activity in obese vs non-obese women. Ten of 13 s-P increases occurred on day 8 and 3 on day 11. All s-P values on day 22 were less than 0.4 ng/mL.

Number of Subjects With Increased S-Progesterone in First and Second Half of Cycle

		Cycle 2 n/N	Cycle 3 n/N
Increase before Day 15	Non-Obese	2/20	5/20
	Obese	2/7	4/6
Increase after Day 15	Non-Obese	0/20	0/20
	Obese	0/7	0/6

Conclusions: Serum progesterone 3 ng/mL or more was observed more often in obese than in non-obese women and mainly limited to week 1 of treatment in cycles 2 and 3.

Background

There are conflicting data regarding a possible link between oral combination contraceptive (COC) failure and body mass index (BMI). Two studies have reported that heavier women were more likely to report an unintended pregnancy during OC use than women who weighed less.[1,2] However, most other studies have reported little or no association between obesity and OC failure.[3,4] For example, no association between BMI and OC failure was found in a clinical trial of 2185 US women, one quarter of whom were obese.[5] Furthermore, no correlation between ovulation rates and BMI was found in subjects who were compliant users of OCs. However, a statistically significant difference was found in compliance in that a larger proportion of obese women (28%) were noncompliant compared with non-obese women (9.7%).[6]

Here we present data regarding elevated serum progesterone values indicative of luteal activity or possible ovulation in obese versus non-obese women treated with a novel combination low-estrogen dose skin patch, AG200-15, in a phase 2 clinical trial. Safety, ovulation inhibition, and pharmacokinetic data from this trial have been previously reported.[7]

Objective

Evaluate pattern of increase in plasma progesterone levels in obese and non-obese healthy women treated with AG200-15, a weekly combination transdermal contraceptive delivery system containing levonorgestrel (LNG) and ethinyl estradiol (EE).

Methods

Study Design

- The contraceptive patch, AG200-15, delivering 100-120 μg/d LNG with 25-30 μg/d EE was evaluated in a multicenter, open-label, single-arm extension trial.
- After providing informed consent, eligible subjects received AG200-15 for 3 cycles (Figure 1).
- Enrolled subjects applied the contraceptive patch to the lower abdomen on the morning of Cycle Day 1 (Day 1 of their menstrual cycle).
- The patch was replaced on Cycle Days 8 and 15, and removed without being replaced on the morning of Cycle Day 22.
- There were no BMI restrictions for enrollment.

Figure 1. Trial Design

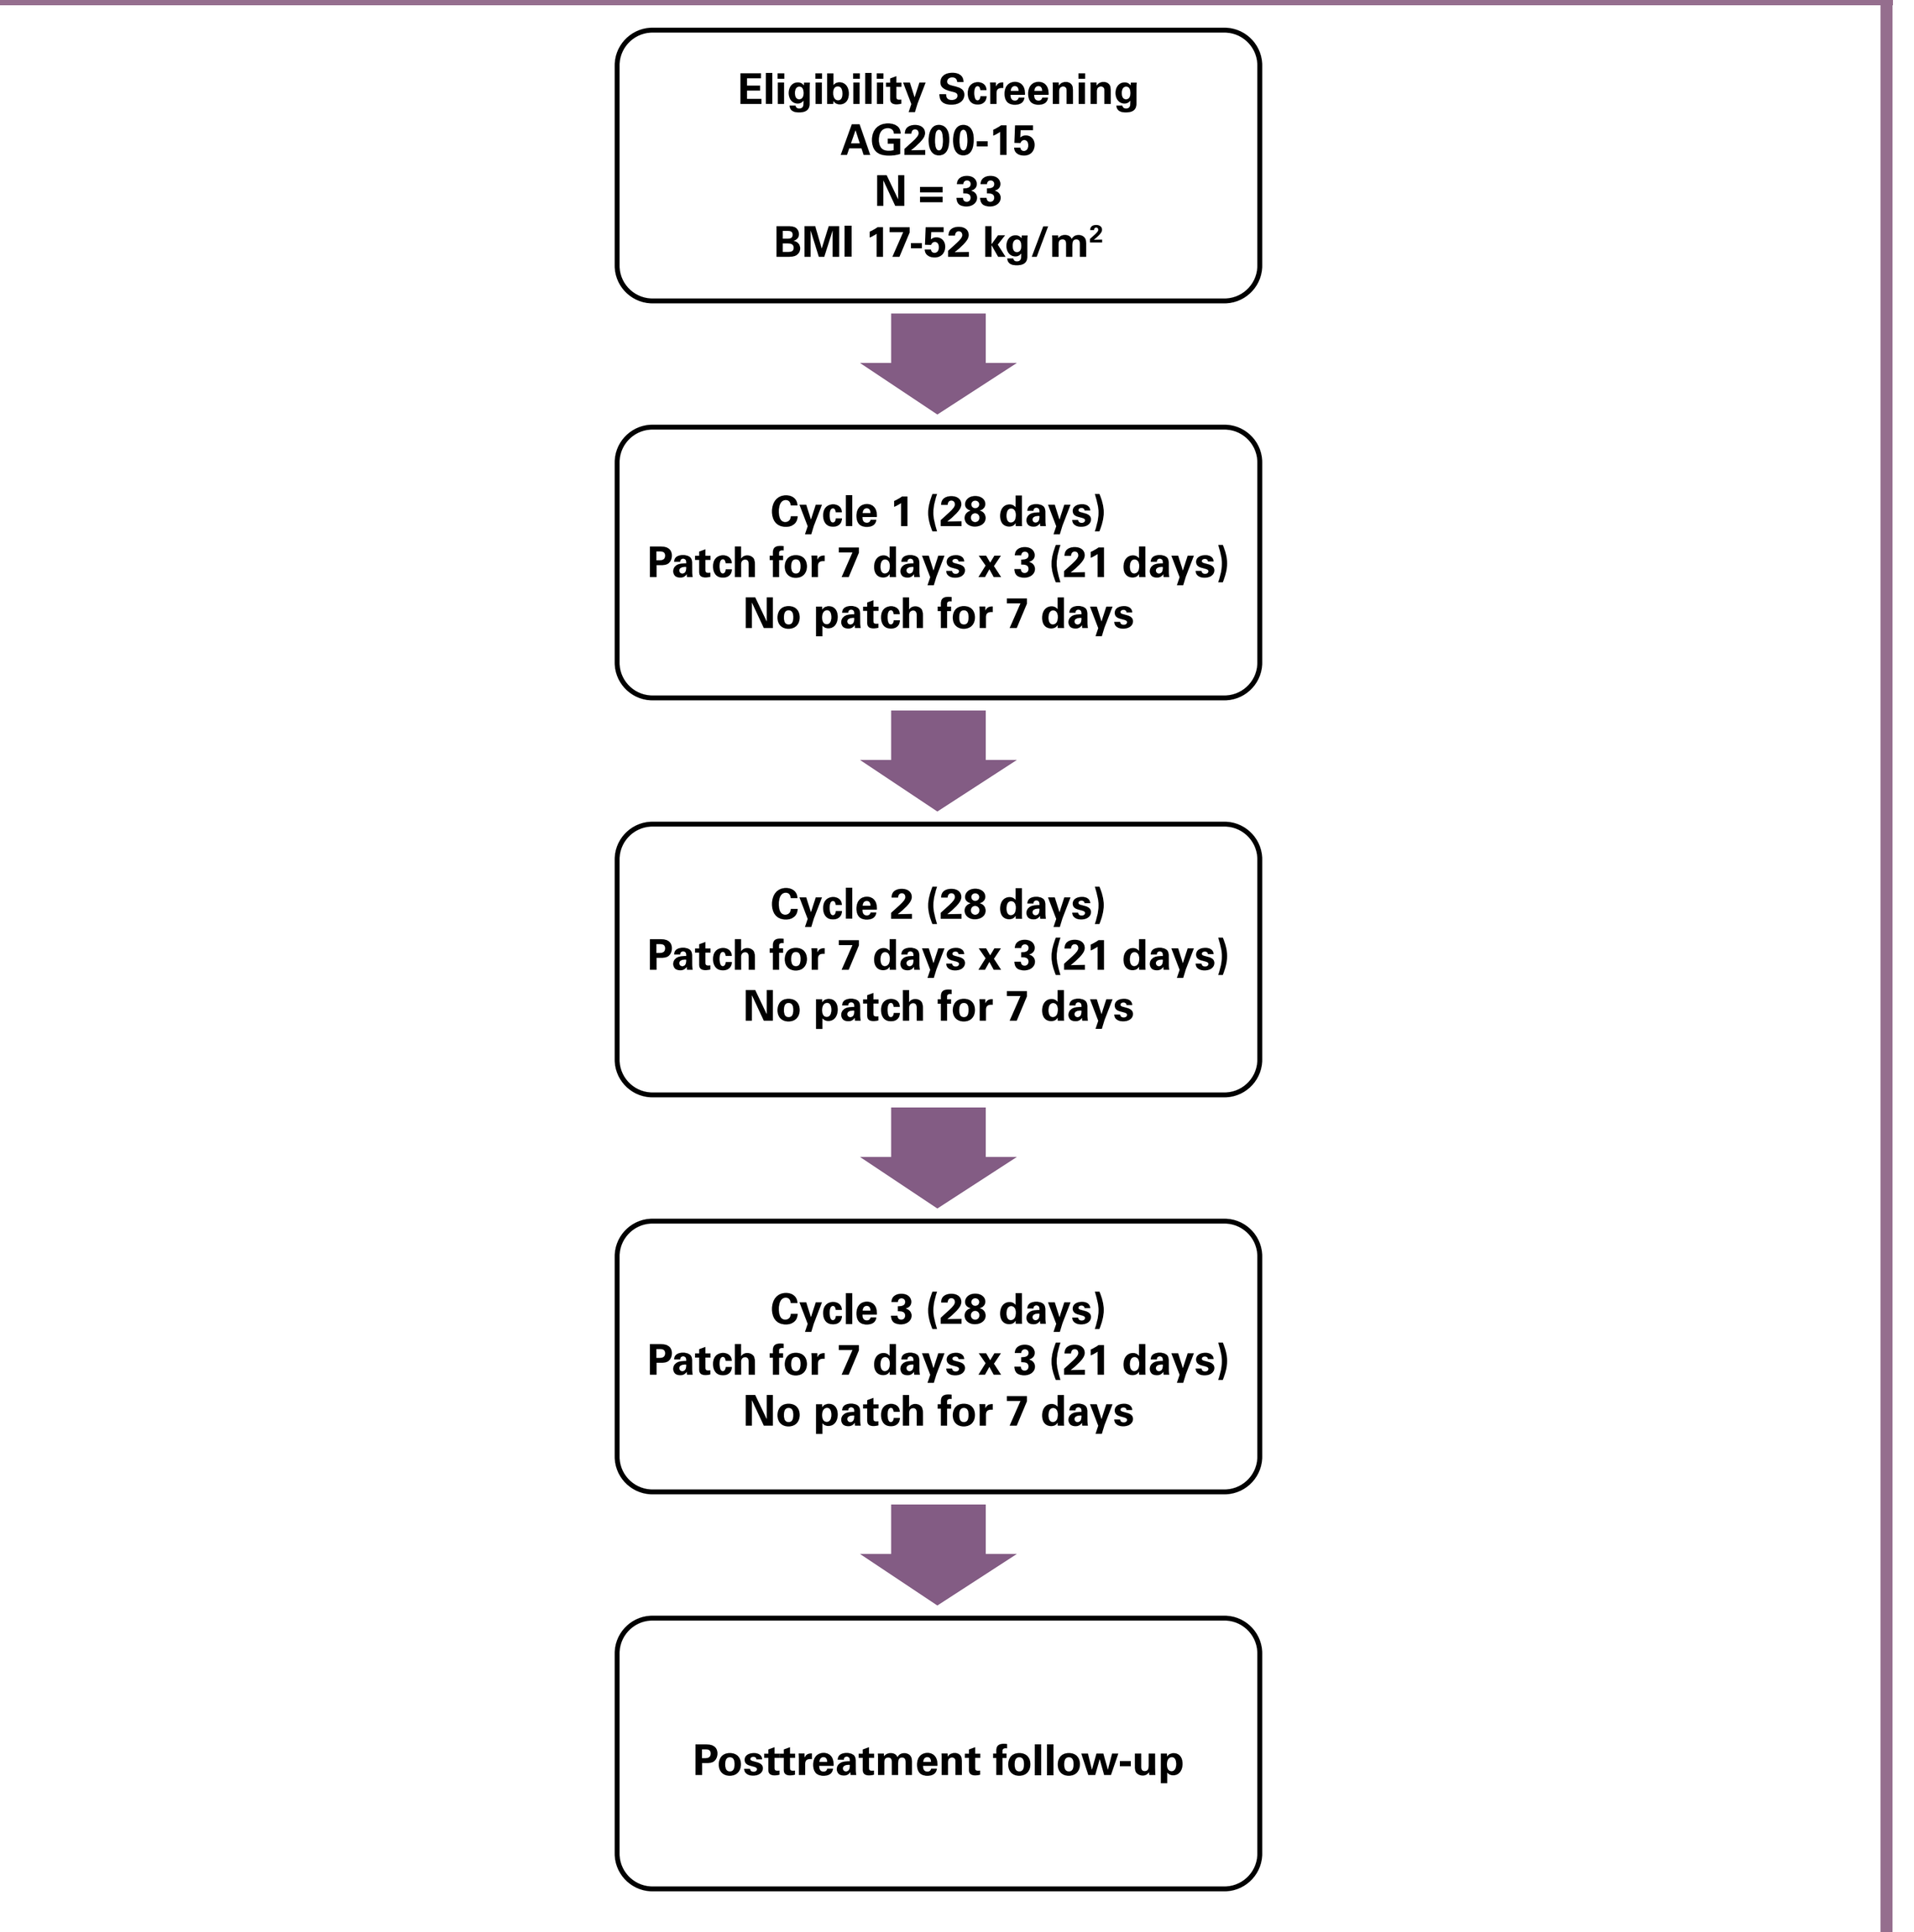
Eligibility Screening
AG200-15
N = 33
BMI 17-52 kg/m²
Cycle 1 (28 days)
Patch for 7 days x 3 (21 days)
No patch for 7 days
Cycle 2 (28 days)
Patch for 7 days x 3 (21 days)
No patch for 7 days
Cycle 3 (28 days)
Patch for 7 days x 3 (21 days)
No patch for 7 days
Posttreatment follow-up

Blood Sampling for Progesterone Evaluations

- Blood samples were drawn for progesterone, a marker of ovulation suppression, on Cycle Days 1, 8, 11, 15, 19, 22, and 25, during each cycle of transdermal patch use.
- Plasma concentrations of progesterone were determined via a validated liquid chromatography–mass spectrometry method.
- The first cycle day of increased serum progesterone levels (serum progesterone ≥3 ng/mL) was determined for obese and non-obese subjects during Cycles 1 to 3.

Results

Demographics and Disposition

- A total of 33 subjects were treated with AG200-15; 8 (24%) were obese (BMI ≥30 kg/m^2).
- Subject demographics for obese and non-obese subjects are summarized in Table 1.

Table 1. Demographics of Subjects Treated With AG200-15

Parameter	Non-Obese (BMI <30 kg/m^2) n=25	Obese (BMI ≥30 kg/m^2) n=8
Age, y		
Mean (SD)	31.9 (8.4)	34.5 (5.2)
Median (range)	33 (18-43)	35 (28-43)
BMI, kg/m^2		
Mean (SD)	24.7 (3.0)	38.5 (6.5)
Median (range)	24.7 (16.8-28.8)	36.8 (32.2-52.4)
Race, n (%)		
Caucasian	13 (52)	5 (62.5)
Black	10 (40)	3 (37.5)
Other	2 (8)	0

Progesterone Levels/Ovarian Suppression in Non-Obese and Obese Subjects

- Progesterone data from 30 subjects were evaluable for comparative evaluation of ovarian suppression in non-obese and obese women.
- Mean progesterone values over all evaluated treatment days for each cycle are presented in Table 2.

Table 2. Mean Progesterone Values for Obese vs Non-Obese Subjects Cycles 1-3

	Cycle 1		Cycle 2		Cycle 3	
	Non-Obese	Obese	Non-Obese	Obese	Non-Obese	Obese
n	23	7	20	7	20	6
Mean progesterone level, ng/mL (SD)	0.57 (1.07)	0.71 (0.77)	0.46 (0.58)	0.77 (1.20)	1.18 (1.66)	1.97 (1.81)

- There was a trend for higher serum progesterone levels in obese subjects versus non-obese subjects. However, the difference was not statistically significant.
- Figure 2 summarizes the first cycle day on which progesterone values exceeded 3.0 ng/mL for Cycles 1, 2, and 3.

Figure 2. Percentage of Subjects With First Cycle Day of Progesterone ≥3 ng/mL

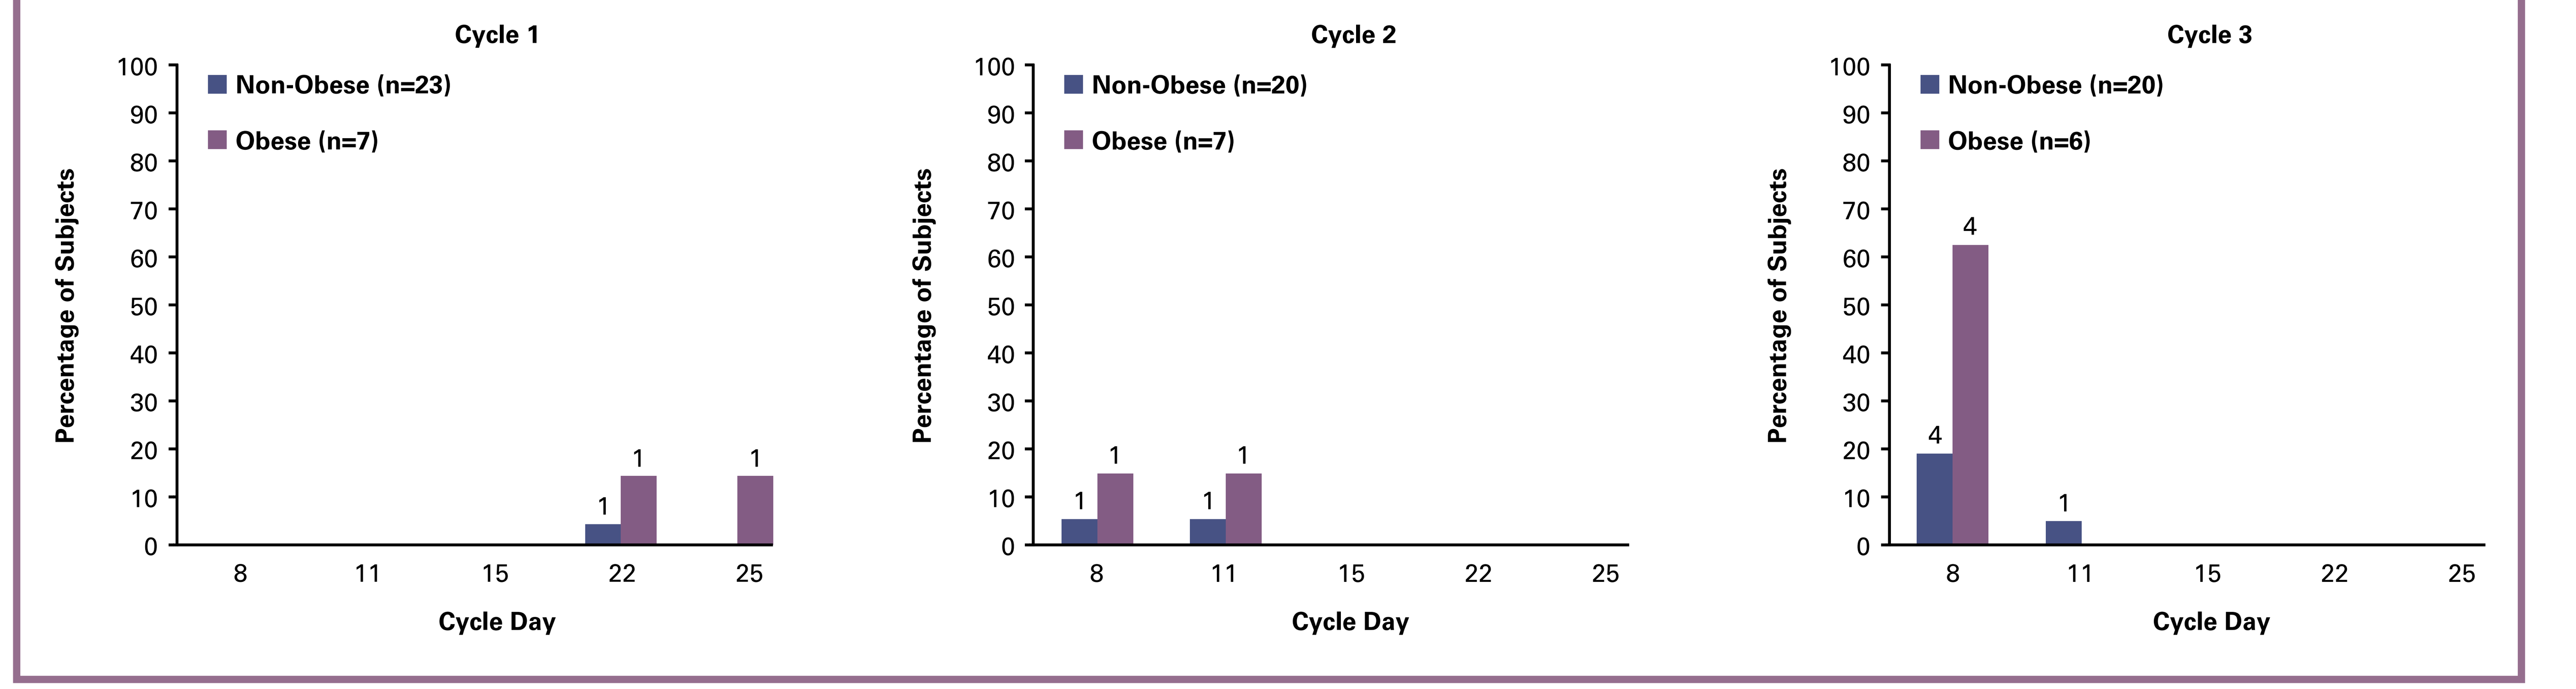
Cycle 1
Non-Obese (n=23)
Obese (n=7)
Percentage of Subjects
100
90
80
70
60
50
40
30
20
10
0
1
1
1
8
11
15
22
25
Cycle Day
Cycle 2
Non-Obese (n=20)
Obese (n=7)
Percentage of Subjects
1
1
1
1
8
11
15
22
25
Cycle Day
Cycle 3
Non-Obese (n=20)
Obese (n=6)
Percentage of Subjects
4
4
1
8
11
15
22
25
Cycle Day

Figure 2. Percentage of non-obese and obese subjects with first cycle day of progesterone ≥3 ng/mL during Cycles 1, 2, and 3 of treatment with AG200-15. Numbers above the bars indicate the number of patients who had progesterone ≥3 ng/mL on that cycle day.

- Table 3 summarizes the percentage and number of non-obese and obese subjects with increased serum progesterone values occurring in the first half (before Day 15) and second half (after Day 15) of each treatment cycle.

Table 3. Percentage of Subjects With Serum Progesterone ≥3 ng/mL in First and Second Half of Each Cycle

		Cycle 1 % (n/N)	Cycle 2 % (n/N)	Cycle 3 % (n/N)
Increase before Day 15	Non-Obese	0 (0/23)	10 (2/20)	25 (5/20)
	Obese	0 (0/7)	29 (2/7)	67 (4/6)*
Increase after Day 15	Non-Obese	4 (1/23)	0 (0/20)	0 (0/20)
	Obese	29 (2/7)	0 (0/7)	0 (0/6)

*Significantly different from non-obese; *P*=0.043, Fisher's exact test.

- The first cycle day of increased serum progesterone levels (≥3 ng/mL) occurred most frequently on Day 8 and before Day 15 in Cycles 2 and 3 in both obese and non-obese subjects (Figure 2, Table 3).
- All serum progesterone values were <3 ng/mL from Day 15 until the end of the cycle, with the exception of 1 subject in whom the increase lasted from Day 8 through Day 19 of the cycle.
- Serum progesterone of ≥3 ng/mL occurred more frequently in obese than in non-obese subjects. No luteal activity was seen on Days 22 and 25 in Cycles 2 and 3.

Discussion

- These observations suggest that obese women experience more luteal activity early in the cycle compared with non-obese women, but that ovulation is not different in Cycles 2 and 3.
- This is in accordance with findings by Westhoff et al.[6] These investigators determined ovulation by ultrasound, but did not measure blood progesterone values until the last 2 weeks of the cycle (Hoogland Score[8]).
- None of the progesterone levels were increased in the normal luteal phase of the cycle for Cycles 2 and 3.
- All increased progesterone levels occurred in the normal follicular phase of the cycle for Cycles 2 and 3.
- The increased luteal activity in obese women observed in the current study may be related to less compliance as found by Westhoff et al[6] or to the observation that there is a negative correlation between LNG levels and BMI.
- Lack of the progesterone pattern associated with ovulation in Cycles 2 and 3 suggest a similar contraceptive effect in obese and non-obese women.
- A phase 3 study that addresses pregnancy risk in obese and non-obese women using the AG200-15 patch is currently ongoing.

Conclusions

- The likelihood of luteinization may be higher in obese subjects using the contraceptive skin patch AG200-15 compared with non-obese patients.
- There is no difference in ovulation between obese and non-obese women based on lack of increased serum progesterone levels beyond Cycle Day 15.
- These findings are limited by sample size and further evaluation is necessary.

References

1. Holt VL, Cushing-Haugen KL, Daling JR. Body weight and risk of oral contraceptive failure. *Obstet Gynecol.* 2002;99:820-827. **2.** Holt VL, Scholes D, Wicklund KG, Cushing-Haugen KL, Daling JR. Body mass index, weight, and oral contraceptive failure risk. *Obstet Gynecol.* 2005;105:46-52. **3.** Trussell J, Schwarz EB, Guthrie K. Obesity and oral contraceptive pill failure. *Contraception.* 2009;79:334-338. **4.** Kaneshiro B, Edelman A, Carlson N, Nichols M, Jensen J. The relationship between body mass index and unintended pregnancy: results from the 2002 National Survey of Family Growth. *Contraception.* 2008;77:234-238. **5.** Westhoff C, Reape K, Shu H. Impact of body weight on observed pregnancy rates with a low-dose estrogen, 91-day extended regimen oral contraceptive. *Contraception.* 2009;80: 196-197. **6.** Westhoff CL, Torgal AH, Mayeda ER, Stanczyk FZ, Lerner JP, Benn EK, Paik M. Ovarian suppression in normal-weight and obese women during oral contraceptive use: a randomized controlled trial. *Obstet Gynecol.* 2010;116(2 Pt 1):275-283. **7.** Stanczyk FZ, Rubin A, Flood L, Foegh M. Pharmacokinetics, tolerability and cycle control of three transdermal contraceptive delivery systems containing different doses of ethinyl estradiol and levonorgestrel. *Horm Mol Biol Clin Invest.* 2011;6:231-240. **8.** Hoogland HJ, Skouby SO. Ultrasound evaluation of ovarian activity under oral contraceptives. *Contraception.* 1993;47:583-590.

This study was sponsored by Agile Therapeutics, Inc.

Poster presented at the American College of Obstetricians & Gynecologists 59th Annual Clinical Meeting, April 30-May 4, 2011, Washington, D.C.